Characterization of Histologic Patterns of Improvement Following Treatment With Efruxifermin (EFX) in NASH Patients With Fibrosis

Cynthia A. Behling[1], Erik J. Tillman[2], Reshma Shringarpure[2], Brittany De Temple[2], Erica Fong[2], Andrew Cheng[2], Timothy P Rolph[2], Stephen A. Harrison[3], and Kitty Yale[2]

(1) Pacific Rim Pathology, San Diego, CA, USA (2) Akero Therapeutics, South San Francisco, CA, USA (3) Pinnacle Clinical Research, San Antonio, TX, USA

AASLD Nov. 12-15, 2021 The Liver Meeting® DIGITAL EXPERIENCE

INTRODUCTION

- Efruxifermin (EFX) is a long-acting Fc-FGF21 analogue being developed as a potential therapy for patients with NASH and fibrosis.
- In the phase 2a BALANCED study[1] in patients with biopsy-confirmed NASH (F1-3), EFX treatment resulted in:
 - 68% -72% relative reduction in liver fat content (LFC) at Week 12
 - normalization of liver fat (<5%) in 48% of patients
 - Improvements in liver histology following 16 weeks of treatment:
 - Fibrosis improvement by ≥1 stage in 48% of patients
 - NASH resolution in 48%
 - Improvement in Non-alcoholic Fatty Liver Disease (NAFLD) Activity Score (NAS) by ≥2 in 58%

AIM

- The aim of this analysis was to better characterize the rapid resolution of NASH and fibrosis by noting qualitative differences in histopathology, and any association of these differences with clinical markers of liver injury, hepatocyte stress, glucose metabolism and fibrosis, as well as patients' PNPLA3 genotype.

METHODS

- At baseline, all patients had biopsy confirmed NASH (F1-F3) with NAS ≥4.
- End-of-Study biopsies were available for 40 EFX-treated patients, all of whom achieved ≥30% relative reduction in LFC on MRI-PDFF.
- All pre- and post-treatment biopsies were reviewed by a single pathologist blinded to treatment and sequence.
- Post-hoc analysis of the recorded histopathology revealed distinct patterns of resolution which were classified qualitatively:
 - 4 categories based on extent and features of steatohepatitis (*see Figure 1*)
 - 3 categories based on extent and features of fibrosis regression (*see Figure 4*)
 - Commonly accepted pathologic findings of hepatic fibrosis regression such as interrupted septa or isolated, chunky collagen were recorded as present or absent.
- Associations* between extent of resolution of histopathology with clinical and genetic biomarkers at baseline and/or following treatment with EFX for up to 16 weeks were evaluated.

**All analyses are qualitative with no hypothesis testing or inferential statistics due to small group sizes*

RESULTS: STEATOHEPATITIS RESOLUTION CATEGORIES

Table 1. Baseline Characteristics by Extent of Steatohepatitis Resolution

EFX-treated patients with end-of-study biopsy (N=40)	NASH or Ballooning Resolution* N=23 (58%)	Steatosis Resolution with Persistent Ballooning N=7 (18%)	Minimal (Residual) NASH N=5 (13%)	Persistent NASH N=5 (13%)
Description	Ballooning = 0, Steatosis = any, Lobular inflammation = any	Ballooning = 1 or 2, Steatosis = 0, Lobular inflammation = any	Ballooning = 1, Steatosis = 1, Lobular inflammation = 1	NAS ≥4, ≥1 point for ballooning, steatosis, and lobular inflammation
Age	50.6	55.4	57.2	54.4
Females, n (%)	10 (43%)	5 (71%)	2 (40%)	3 (60%)
Hispanic or Latino, n (%)	16 (70%)	3 (43%)	1 (20%)	4 (80%)
NAS	5.4	5.3	5.8	6.4
LFC %	18.7	14.9	22.4	24.8
Type 2 Diabetes, n/N (%)	8/23 (35%)	7/7 (100%)	1/5 (20%)	2/5 (40%)
HbA1c %	6.00	7.47	6.52	6.1
BL F1/F2/F3	10/7/6	3/2/2	2/2/1	3/0/2
Patients genotyped for PNPLA3 (N)	*22*	*6*	*4*	*5*
Patients with 0 risk alleles, n (%)	4 (18%)	3 (50%)	2 (50%)	1 (20%)
Patients with 1 risk allele, n (%)	9 (41%)	3 (50%)	2 (50%)	4 (80%)
Patients with 2 risk alleles, n (%)	9 (41%)	0	0	0
Median ALT, U/L	46.5	52.5	44.5	69.5
Median AST, U/L	31.5	40.5	34.0	35.0
Median GGT, U/L	42	60	40	72
Median Pro-C3 µg/mL	13.7	16.0	13.6	17.1
Median ELF score	9.17	10.21	9.68	9.81

*All patients in this category had ballooning resolution and 19 of 23 had NASH resolution. Demographics and characteristics are presented as means unless otherwise noted.

Table 2. Histological Outcomes and Improvements in Biomarkers of Liver Health Following EFX Treatment by Categories of Steatohepatitis Resolution

EFX-treated patients with end-of-study biopsy (N=40)	NASH or Ballooning Resolution* N=23 (58%)	Steatosis Resolution with Persistent Ballooning; N=7 (18%)	Minimal (Residual) NASH N=5 (13%)	Persistent NASH N=5 (13%)
Description	Ballooning = 0, Steatosis = any, Lobular inflammation = any	Ballooning = 1 or 2, Steatosis = 0, Lobular inflammation = any	Ballooning = 1, Steatosis = 1, Lobular inflammation = 1	NAS ≥4, ≥1 point for ballooning, steatosis, and lobular inflammation
LFC normalized (≤5% LFC), n (%)	13 (56.5%)	7 (100%)	0 (0%)	1 (20%)
NAS (mean CFB)	Improved (-3.7)	Improved (-3.3)	Improved (-2.8)	Unchanged (-1.6)
NASH resolution, n (%)	19 (83%)	0 (0%)	0 (0%)	0 (0%)
≥1- / 2-stage Fibrosis improvement	14 / 8	3 / 1	3 / 1	2 / 1
Fibrosis improvement + NASH resolution	11	0	0	0
ALT, median % CFB	-50.5%	-68.4%	-30.3%	-59.7%
AST, median % CFB	-39.5%	-61.3%	-48.6%	-55.9%
GGT, median % CFB	-41.9%	-48.5%	-26.2%	-28.6%
Pro-C3, median µg/mL CFB	-3.9	-4.2	-5.7	-7.2
ELF score, median absolute CFB	-0.51	-0.97	-0.40	-0.56

*All patients in this category had ballooning resolution and 19 of 23 had NASH resolution. Demographics and characteristics are presented as means unless otherwise noted.

Figure 1. Characteristics of Steatohepatitis

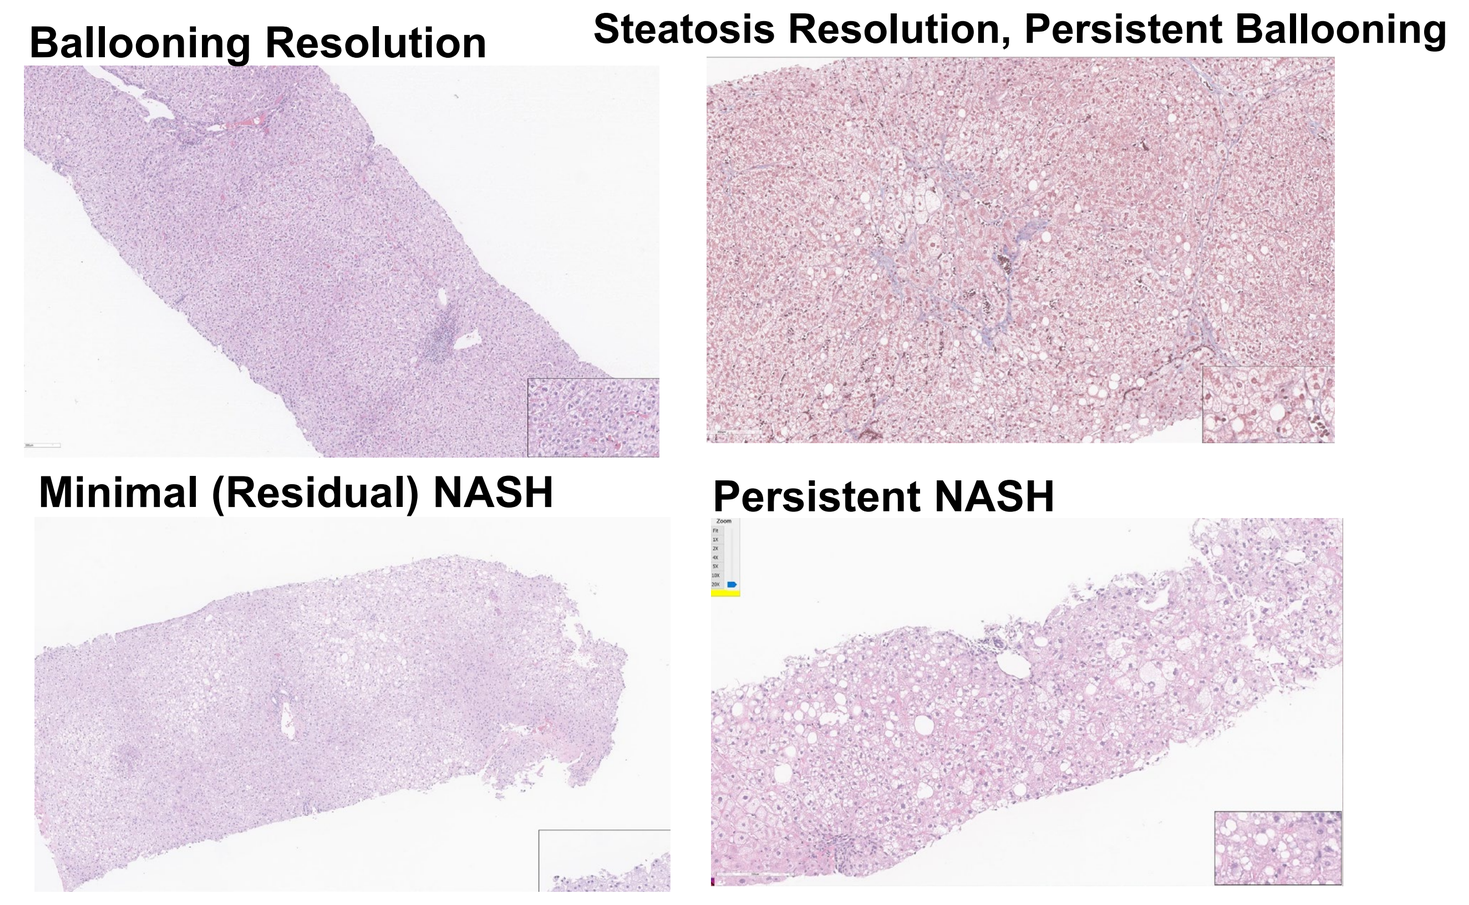
Ballooning Resolution
Steatosis Resolution, Persistent Ballooning
Minimal (Residual) NASH
Persistent NASH

Figure 2. Patients With Steatosis Resolution and Persistent Ballooning Were More Insulin Resistant at Baseline.

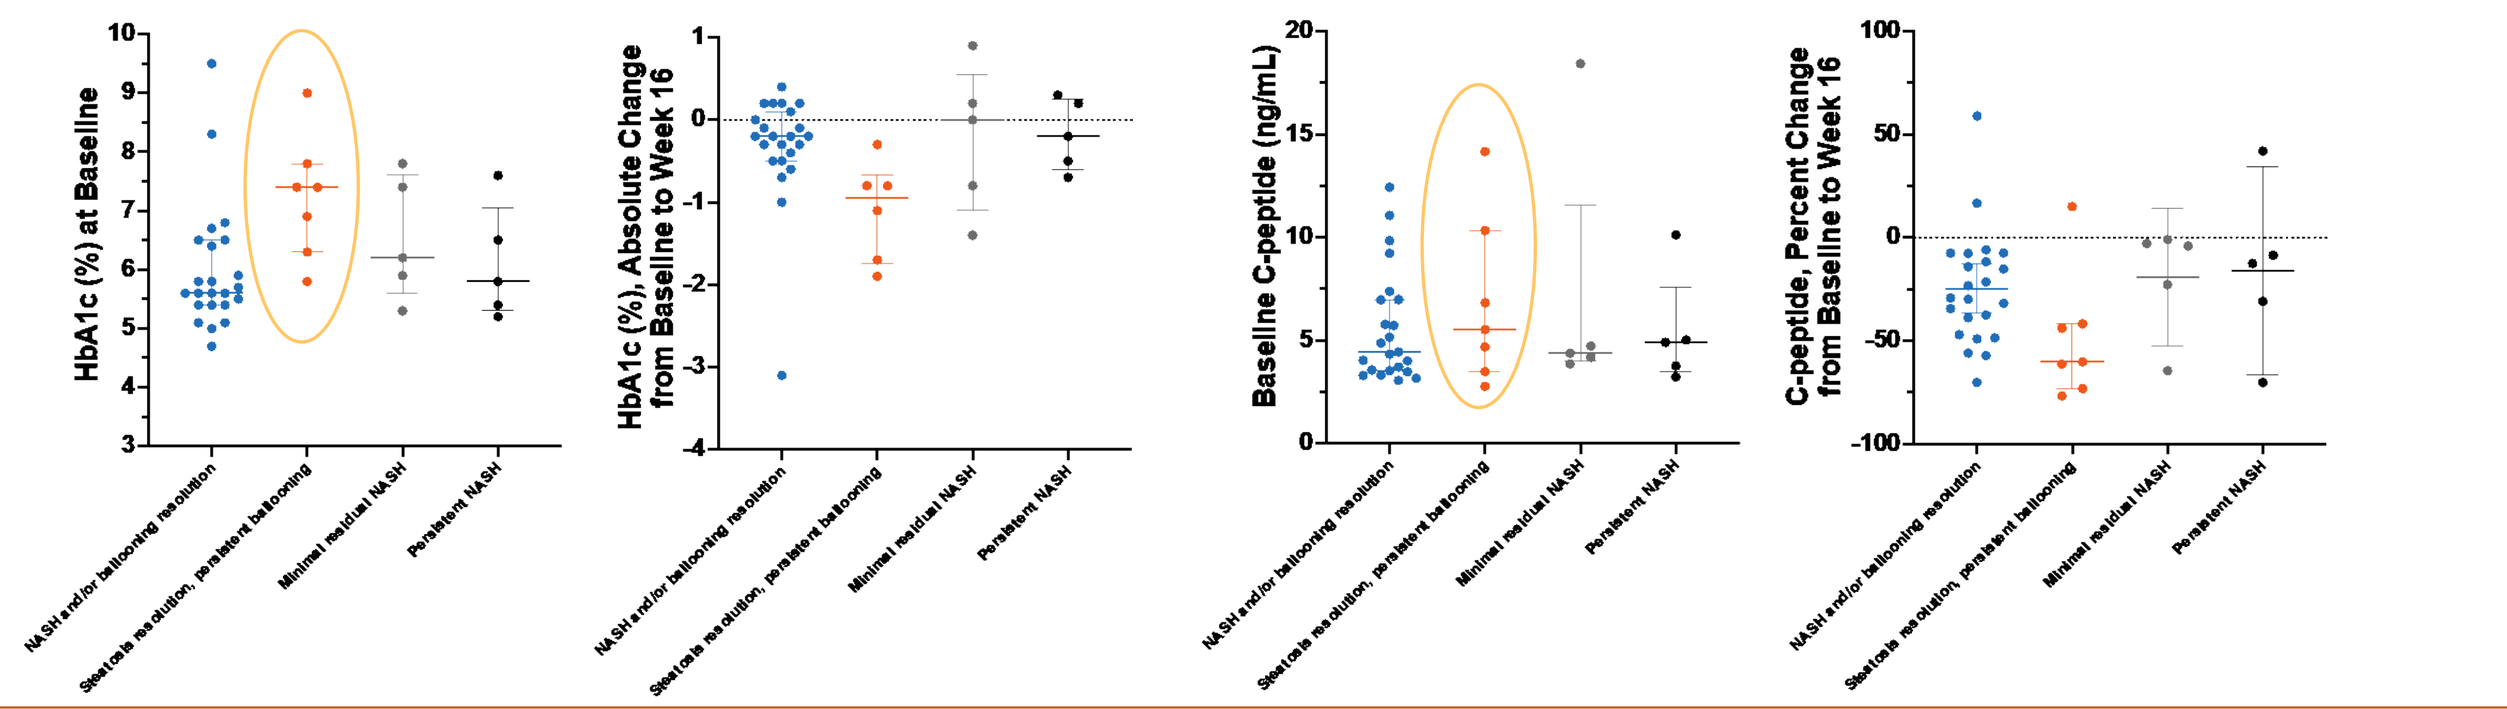
HbA1c (%) at Baseline
HbA1c (%), Absolute Change from Baseline to Week 16
Baseline C-peptide (ng/mL)
C-peptide, Percent Change from Baseline to Week 16
NASH and/or ballooning resolution
Steatosis resolution, persistent ballooning
Minimal residual NASH
Persistent NASH

- This pattern is consistent with hepatocyte ballooning being more prevalent in patients with diabetes.[2,3]
- 100% of patients in this category had type 2 diabetes.
- Improvement in steatosis was associated with insulin sensitizing actions of EFX.
- A longer treatment duration (>16 weeks) may be required to translate improved metabolic health into ballooning resolution.

Lines and bars represent Median ± IQR

Figure 3. Patients With Persistent Ballooning had More Advanced Liver Disease at Baseline but Greater improvements in Markers of Liver Injury and Fibrosis with EFX

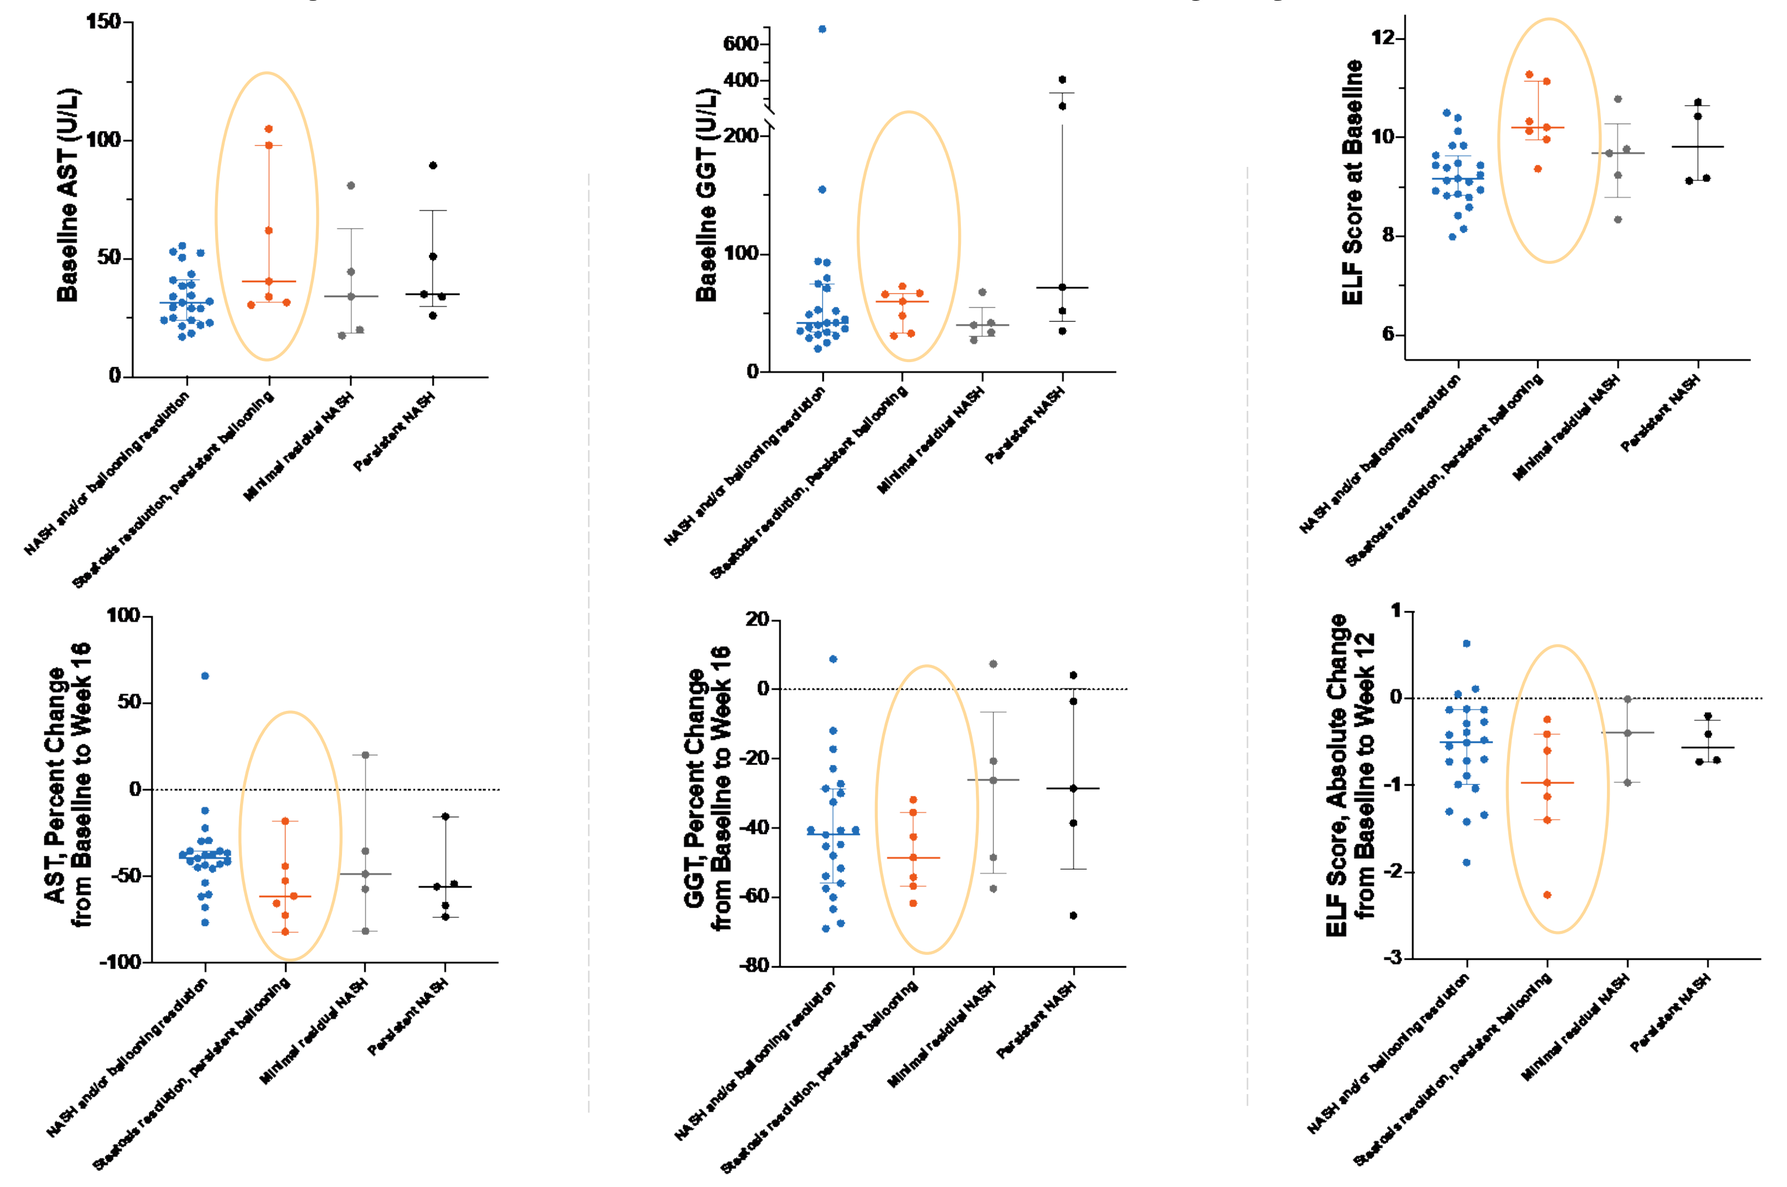
Baseline AST (U/L)
Baseline GGT (U/L)
ELF Score at Baseline
AST, Percent Change from Baseline to Week 16
GGT, Percent Change from Baseline to Week 16
ELF Score, Absolute Change from Baseline to Week 12

Lines and bars represent Median ± IQR

Steatohepatitis Resolution Categories: Summary of Observations

- 87% of EFX treated patients (35 of 40) showed improvement in steatohepatitis after only 16 weeks of treatment.
 - Almost half (48%) of all EFX-treated patients achieved resolution of NASH.
- Across all 4 categories, most subjects achieved improvements in markers of metabolic health, liver injury, hepatocyte stress, and fibrosis.
- Patients at higher risk of NASH progression, e.g. homozygous carriers of the PNPLA3 risk allele, as well as those of Hispanic ethnicity, appeared to be as responsive to EFX as patients at lower risk of progression.
- The categories with less resolution of steatohepatitis, despite comparable improvements in markers of liver injury and fibrosis suggest it may take longer in some patients to achieve complete resolution:
 - Patients in these categories had more severe disease, as indicated by higher LFC and levels of liver injury markers, as well as poorer glycemic control at baseline.
 - Improved glycemic control via insulin sensitization may be required for resolution of ballooning.

RESULTS: FIBROSIS REGRESSION CATEGORIES

Table 3. Baseline Characteristics by Categories of Fibrosis Regression

EFX-treated patients with end-of-study biopsy (N=40)	Histological F stage improvement N=22 (55%)	Features of fibrosis regression observed without improvement in F stage (intra-stage improvement) N= 10 (25%)	No improvement in F stage N=8 (20%)
Description	≥1-stage improvement in fibrosis	Interrupted/irregular septa, hepatocyte regeneration, isolated or chunky collagen	Neither F-stage improvement nor qualitative features of regression observed
Mean age	52.7	57.2	47.4
Females, n (%)	8 (36%)	6 (60%)	6 (75%)
Hispanic or Latino, n (%)	12 (55%)	6 (60%)	6 (75%)
Mean Liver Fat Content (%)	20.0	17.6	19.3
Mean NAS	5.5	5.9	5.3
BL F1/F2/F3 (n)	7/10/5	6/1/3	5/0/3
T2D, n (%)	10 (45%)	5 (50%)	3 (38%)
Median HbA1c, %	5.75	6.8	5.8
Patients genotyped for PNPLA3 (N)	*21 genotyped*	*10 genotyped*	*6 genotyped*
Patients with 0 risk alleles, n (%)	4 (19%)	5 (50%)	1 (17%)
Patients with 1 risk allele, n (%)	10 (47%)	3 (30%)	5 (83%)
Patients with 2 risk alleles, n (%)	7 (33%)	2 (20%)	0
Median ALT, U/L	47.5	57.5	45.8
Median AST, U/L	31.8	41.5	37.5
Median GGT, U/L	41	56	61.5
Median Pro-C3, µg/mL	14.4	16.7	15.1
Median ELF score	9.46	9.44	9.03

Table 4. Histological Outcomes and Improvements in Biomarkers of Liver Health Following EFX Treatment by Categories of Fibrosis Regression

EFX-treated patients with end-of-study biopsy (N=40)	Histological F stage improvement N=22 (55%)	Features of fibrosis regression observed without improvement in F stage (intra-stage improvement) N= 10 (25%)	No improvement in F stage N=8 (20%)
Description	≥1-stage improvement in fibrosis	Interrupted/irregular septa, hepatocyte regeneration, isolated or chunky collagen	Neither F-stage improvement nor qualitative features of regression observed
NAS, mean CFB	-3.0	-3.9	-3.0
Normalized (≤5%) Liver Fat Content, n (%)	9 (41%)	6 (60%)	6 (75%)
≥1- / 2-stage F improvement	22 / 11	0 / 0	0 / 0
NASH resolution, n (%)	11 (50%)	4 (40%)	4 (50%)
ALT, median % CFB	-49.8%	-57.6%	-68.9%
AST, median % CFB	-40.5%	-51.4%	-58.6%
GGT, median % CFB	-38.0%	-51.2%	-45.5%
Pro-C3, median µg/mL CFB	-5.0	-6.5	-3.9
ELF score, median absolute CFB	-0.55 (n=21)	-0.56	-0.56
% HbA1c, median absolute CFB	-0.1	-0.75	-0.5

Figure 4. Histological Patterns of Regression

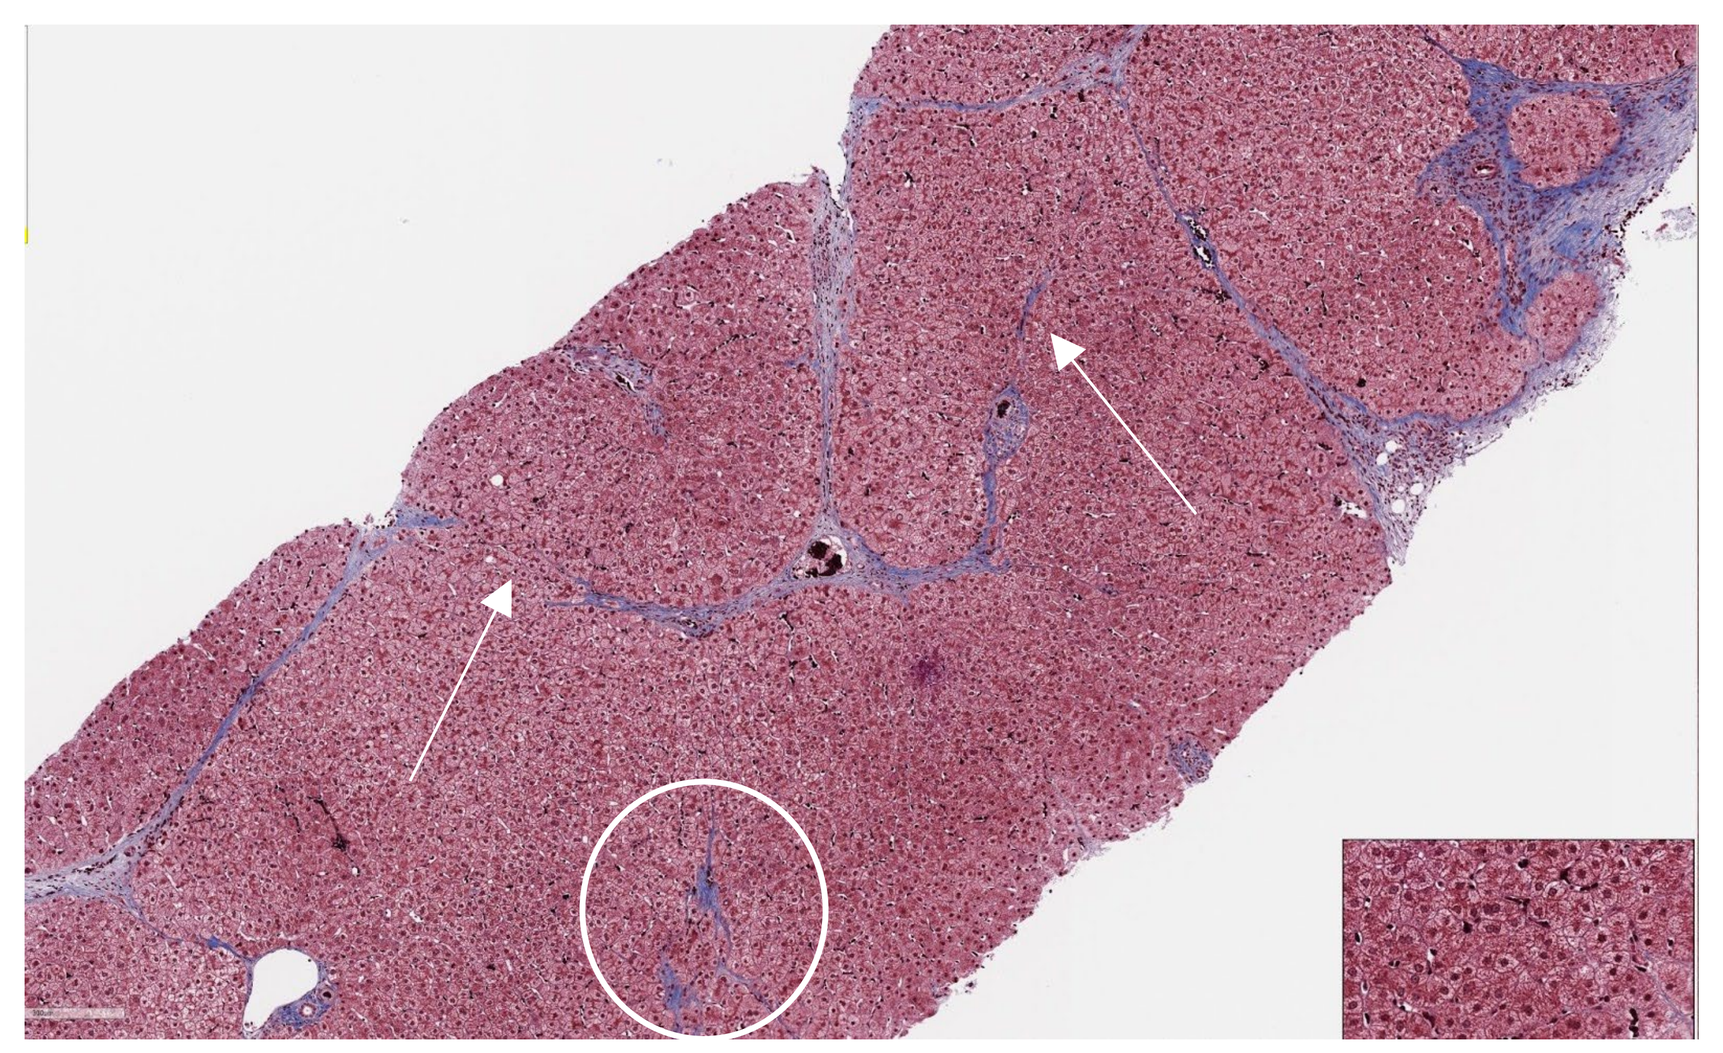

Arrows: Interrupted Septa
Circle: Chunky collagen

Figure 5. Extent of Fibrosis Regression Does Not Correlate With Markers of Liver Injury and Fibrosis at Baseline or After EFX Treatment

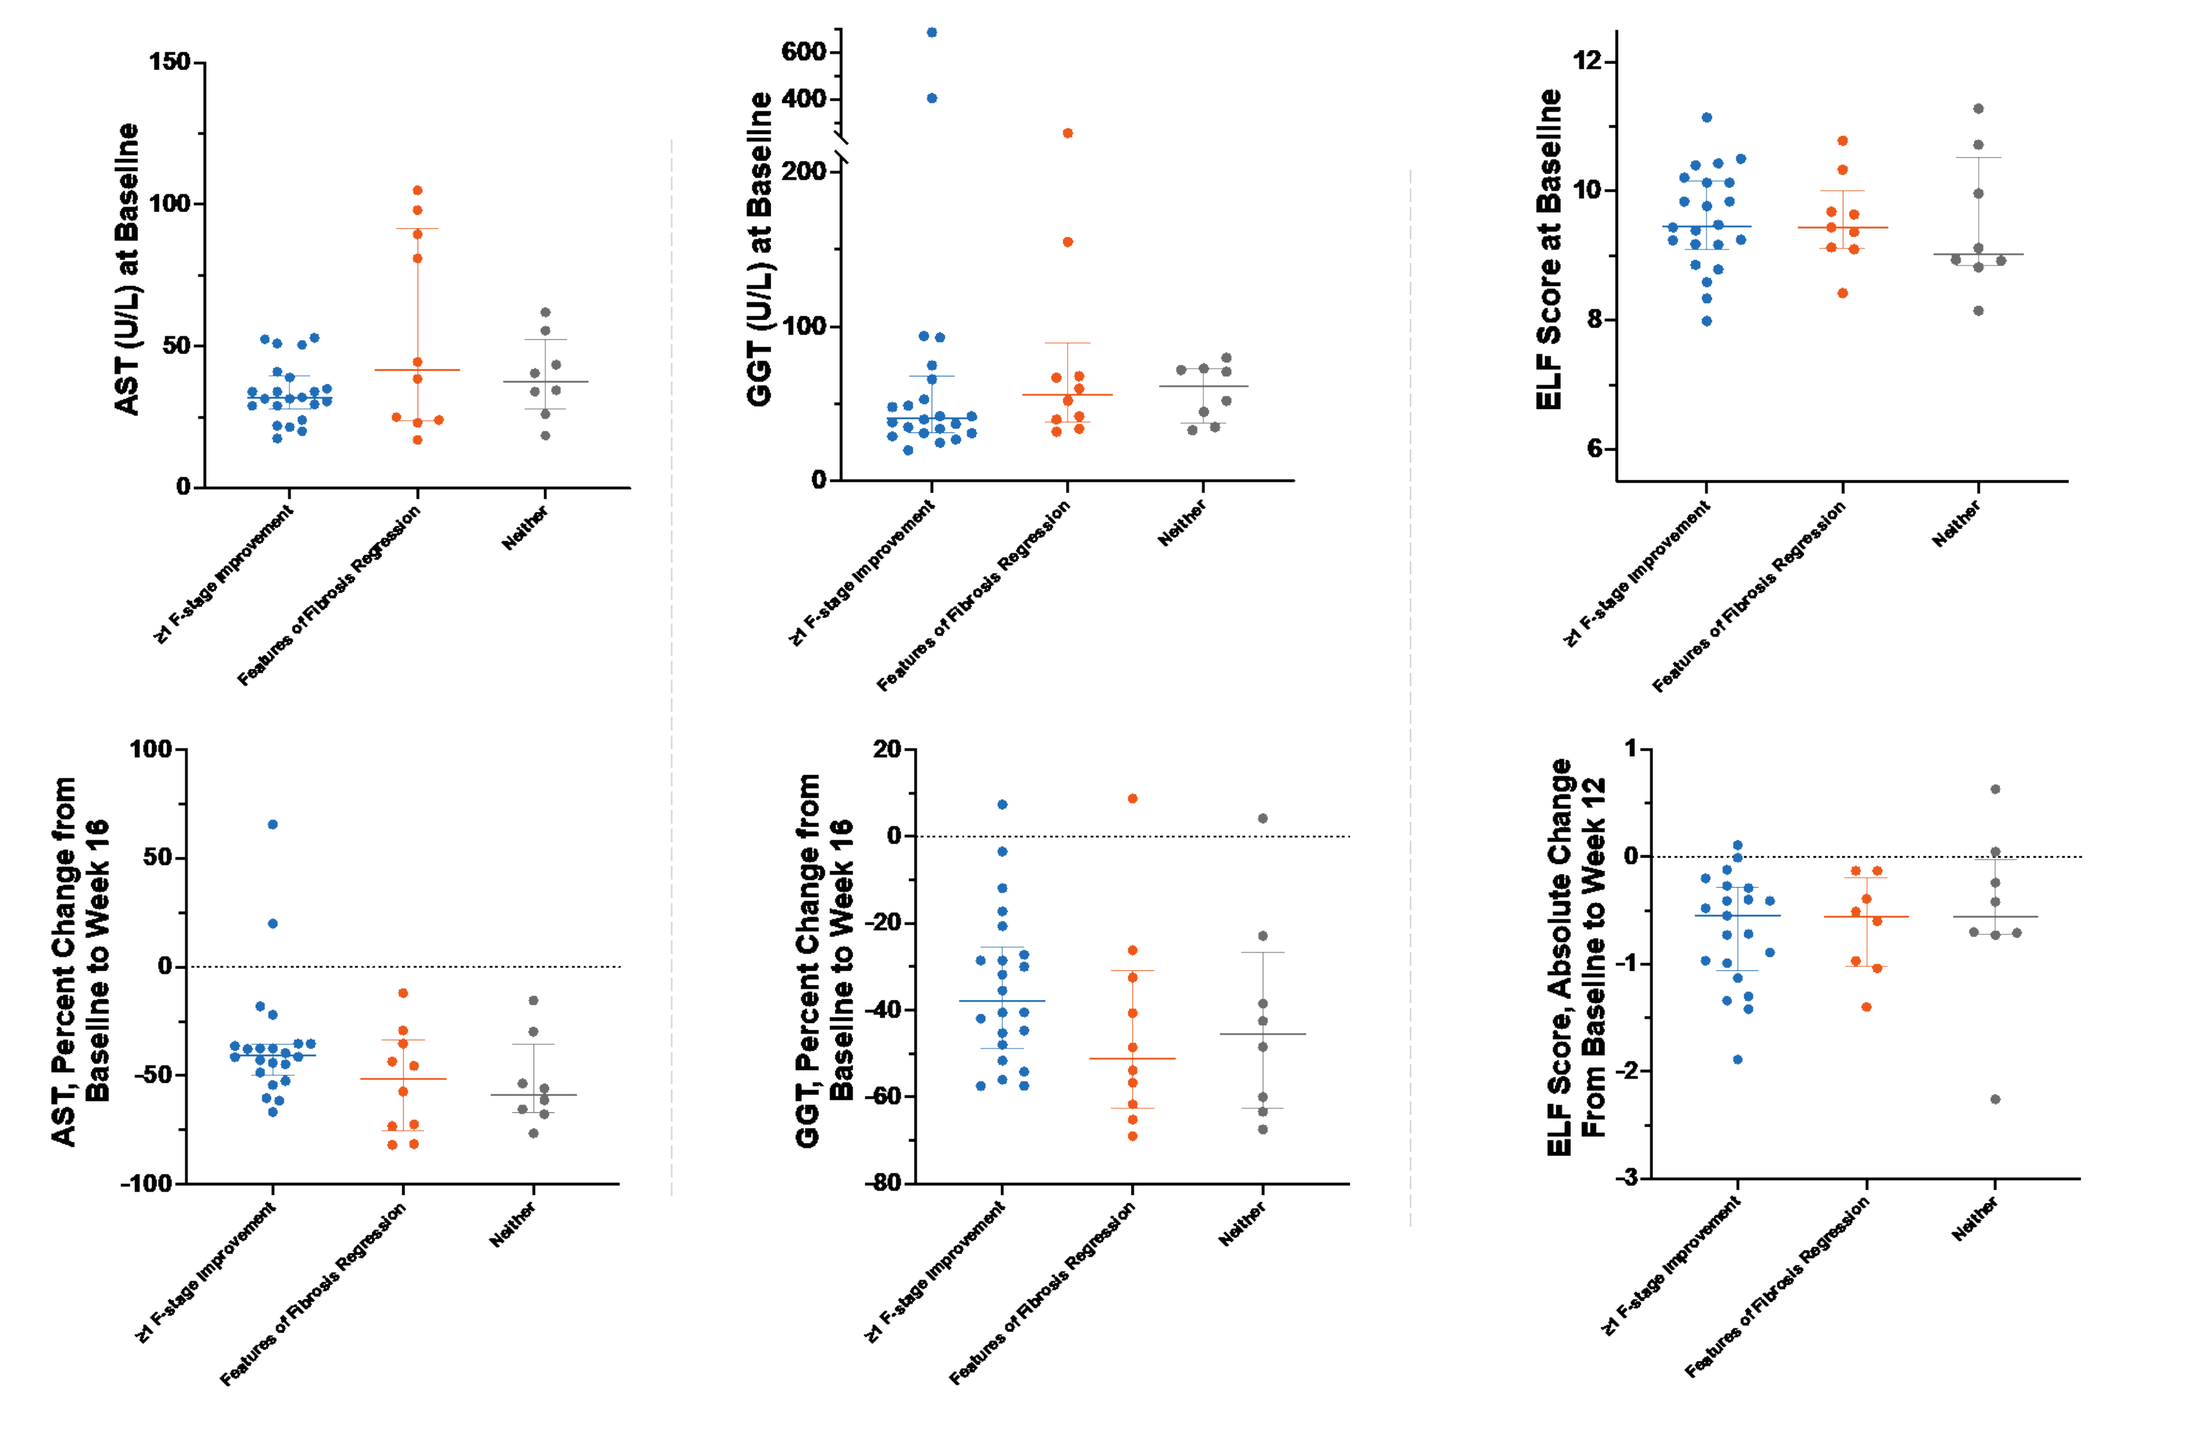
AST (U/L) at Baseline
GGT (U/L) at Baseline
ELF Score at Baseline
AST, Percent Change from Baseline to Week 16
GGT, Percent Change from Baseline to Week 16
ELF Score, Absolute Change From Baseline to Week 12
≥1 F-stage improvement
Features of Fibrosis Regression
Neither

Lines and bars represent Median ± IQR

Fibrosis Regression Categories: Summary of Observations

- 80% of EFX-treated patients (32 of 40) showed qualitative features of fibrosis regression.
 - 55% (22 of 40) patients achieved ≥ 1 stage improvement
 - 50% (11 of 22) of F2/F3 achieved ≥ 2 stages improvement
- The effectiveness of EFX in regressing fibrosis was maintained in patients who were homozygous carriers of the PNPLA3 risk allele who are at a higher risk of fibrosis progression.[4,5]
- The extent of regression of fibrosis did not appear to be dependent on magnitude of liver fat reduction or extent of resolution of steatohepatitis.
 - Suggests EFX has a direct anti-fibrotic effect independent of improvement in LFC and overall metabolic health
- Longer duration of treatment with EFX may be required to see ≥ 1 stage improvement in fibrosis in the group with qualitative features of fibrosis regression, but not a categorical change.
- There were no obvious pretreatment traits, or difference in responsiveness to EFX underlying the group with no discernible signs of fibrosis regression after 16 weeks of EFX treatment.

CONCLUSIONS

- **Most EFX-treated patients with end-of treatment biopsies showed improvements in features of steatohepatitis (35 of 40; 87%) and/or fibrosis (32 of 40; 80%), after only 16 weeks:**
 - **Improvement in histologic characteristics of steatohepatitis, changes in histologic pattern relative to individual features, and intra-stage histologic changes in fibrosis may be early indicators of response after relatively short periods of treatment.**
- **EFX resolved NASH and improved fibrosis by ≥1 stage among patients homozygous for the PNPLA3 allele who are at high risk of progression to cirrhosis.**
- **Evidence of fibrosis improvement without correlation to LFC normalization or NASH resolution is consistent with EFX's potential to have direct anti-fibrotic effects.**
- **Insulin sensitization appears to be an important prerequisite for NASH resolution in patients with poorer overall metabolic health.**
- **Akero's Ph2b studies, HARMONY (F2/3) and SYMMETRY (F4), evaluating improvement in fibrosis after EFX treatment for 24 and 36 weeks, respectively, will determine if longer treatment durations increase response rates by categorical definitions of NASH resolution and fibrosis improvement.**

REFERENCES

1. **Harrison SA, et al.** *Nat Med*. 2021;27(7):1262-1271.
2. **Kakisaka K, et al.** *Sci Rep*. 2021;11(1):15392.
3. **Puchakayala BK, et al.** *World J Hepatol.* 2015;7(25):2610-2618.
4. **Sookoian S, Pirola CJ.** *Hepatology.* 2011 Jun;53(6):1883-94.
5. **Singal AG, et al.** *Am J Gastroenterol.* 2014 Mar;109(3):325-34.

CONTACT INFORMATION

Kitty Yale: Kyale@akerotx.com

ACKNOWLEDGMENTS

The authors wish to thank the patients, their families, and study investigators. This study and all analyses were funded by Akero Therapeutics.

DISCLOSURES

CB's institution receives support for biopsy related work and/ or consulting agreements with Akero Therapeutics. SAH received consulting fees and grant/research support from Akero Therapeutics. EJT, RS, BDT, EF, AC, TPR, and KY are employees of Akero Therapeutics.