Efruxifermin treatment improved histopathology and non-invasive markers of liver injury and fibrogenesis in NASH patients across PNPLA3 genotypes: a *post hoc* analysis of the Ph2a BALANCED study

Erik J Tillman[1], Reshma Shringarpure[1], Guy Neff[2], Rashmee Patil[3], Peter Ruane[4], Bradley Freilich[5], Cynthia Behling[6], Erica Fong[1], Brittany de Temple[1], Kitty Yale[1*], Stephen A Harrison[7], Tim Rolph[1]

[1]Akero Therapeutics, South San Francisco, CA, USA. [2]Covenant Metabolic Specialists, Fort Myers, FL, USA. [3]South Texas Research Institute, Edinburg, TX, USA. [4]Ruane Clinical Research Group, Los Angeles, CA, USA. [5]Kansas City Research Institute, Kansas City, MO, USA. [6]Pacific Rim Pathology, San Diego, CA, USA. [7]Pinnacle Clinical Research, San Antonio, TX, USA.

*Contact: kyale@akerotx.com

BACKGROUND

Efruxifermin (EFX) is a long-acting Fc-FGF21 analogue being developed as a potential therapy for patients with fibrosis due to non-alcoholic steatohepatitis (NASH). In the phase 2a BALANCED study (NCT03976401) of patients with biopsy-confirmed NASH (F1-3), 16-week treatment with EFX significantly reduced liver fat content and improved markers of liver injury, fibrosis, and lipid and glucose metabolism while demonstrating an acceptable safety and tolerability profile[1].

Patatin-like phospholipase domain-containing protein 3 (PNPLA3) is a lipid droplet-associated protein. The I148M variant of PNPLA3 (rs738409) is strongly associated with more severe NAFLD/NASH, and increased risk of progressive fibrosis, cirrhosis, and HCC[2]. Concurrent insulin resistance further increases the risk associated with I148M[3]. Interventions that reverse NASH fibrosis in patients whose aetiology is associated with more severe and progressive disease address the greatest unmet medical need in NASH. The interaction between *PNPLA3* genotype and response to EFX treatment in patients with NASH has not been explored

AIMS

This *post hoc* analysis evaluated the prevalence of *PNPLA3* variants in a patient population, all of whom had biopsy-confirmed F1-F3 NASH. The analysis also evaluated differences in liver and whole-body metabolic characteristics present in I148M heterozygous or homozygous patients with NASH. Finally, the effects of EFX treatment on histopathology, markers of liver health and whole body metabolism were evaluated across *PNPLA3* genotypes.

METHODS

Figure 1. BALANCED Main Study Design

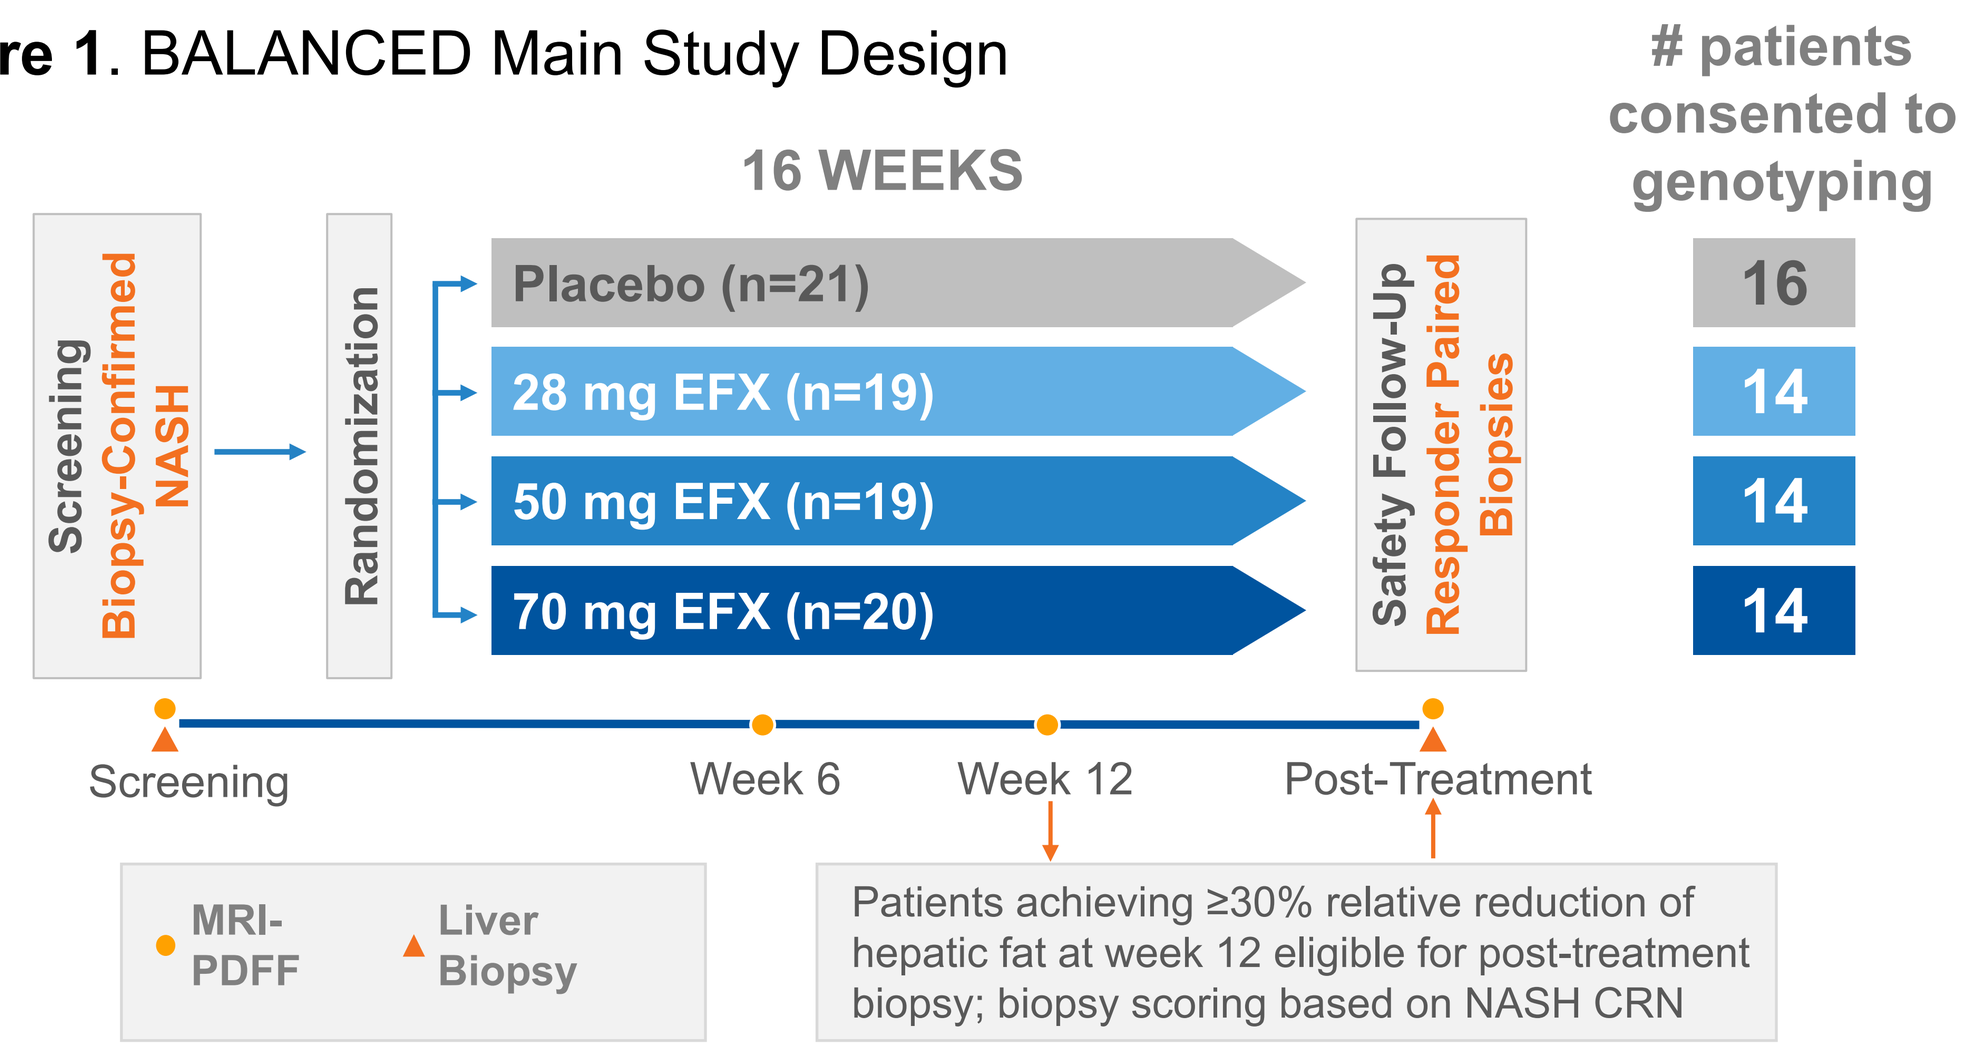

patients consented to genotyping
16 WEEKS
Screening
Biopsy-Confirmed NASH
Randomization
Placebo (n=21)
28 mg EFX (n=19)
50 mg EFX (n=19)
70 mg EFX (n=20)
Safety Follow-Up
Responder Paired Biopsies
16
14
14
14
Screening
Week 6
Week 12
Post-Treatment
MRI-PDFF
Liver Biopsy
Patients achieving ≥30% relative reduction of hepatic fat at week 12 eligible for post-treatment biopsy; biopsy scoring based on NASH CRN

80 patients with biopsy-confirmed F1-F3 NASH were randomized in the BALANCED Main Study (Fig. 1), of whom 79 received at least one dose of study drug. Week 12 MRI-PDFF data were available for 68 patients, and end-of-treatment biopsy was available for 42 patients.

As genotype analysis was a *post hoc* procedure, consent was sought from all study participants. 58 patients consented to undergo genetic analyses.

DNA was extracted from whole blood, and *PNPLA33* rs738409 was genotyped by a validated qPCR method (Medpace). The prevalence of each genotype by dose group is shown in Table 1.

Table 1. Frequency of PNPLA3 genotypes in BALANCED Main Study

Number of patients (% of dose group)	PNPLA3 I148 genotype			No genotype available
	I/I	I/M	M/M	
Placebo	5 (24%)	5 (24%)	6 (29%)	5 (24%)
EFX, all doses	11 (19%)	22 (38%)	9 (16%)	16 (28%)
28 mg EFX	2 (11%)	8 (42%)	4 (21%)	5 (26%)
50 mg EFX	5 (26%)	6 (32%)	3 (16%)	5 (26%)
70 mg EFX	4 (20%)	8 (40%)	2 (10%)	6 (30%)

RESULTS

Table 2. Baseline Demographics and Characteristics

Mean (SD), unless otherwise noted	# of PNPLA3 148M risk alleles			p value*
	0 (I/I)	1 (I/M)	2 (M/M)	
All genotyped patients, N	**16**	**27**	**15**	
Demographics				
Age, years	57.5 (9.9)	51.7 (11.2)	48.5 (14.4)	**0.0377**
Female/male, n	11/5	14/13	7/8	
Hispanic or Latino, n (%)	5 (31%)	17 (63%)	9 (60%)	
T2D, n (%)	10 (63%)	12 (44%)	8 (53%)	
BMI, kg/m^2	35.7 (5.1)	39.4 (7.3)	36.5 (6.1)	0.7079
NASH histology and biomarkers				
Liver fat, MRI-PDFF (%)	17.5 (6.4)	21.3 (8.4)	20.3 (5.9)	0.2786
NAFLD Activity Score	5.13 (1.02)	5.52 (0.94)	5.53 (0.83)	0.2237
F1/F2/F3, n	6/4/6	11/7/9	6/5/4	
Liver injury and fibrosis				
ALT, U/L	55.3 (39.4)	57.4 (33.7)	52.5 (24.7)	0.8203
AST, U/L	44.9 (26.6)	39.4 (18.6)	35.0 (15.1)	0.1815
GGT, U/L	122.4 (164.0)	69.7 (75.6)	52.4 (28.8)	0.0577
ELF score	9.72 (0.97)	9.65 (0.80)	9.30 (0.91)	0.1948
Pro-C3, ng/µL	17.8 (7.5)	18.0 (9.3)	16.6 (6.3)	0.6972
Urate, mg/dL	5.1 (1.1)	6.3 (1.2)	5.7 (1.5)	0.1297
FGF21, pg/mL	638.8 (301.1)	563.7 (484.8)	383.8 (161.0)	0.0669
Glucose metabolism				
Insulin, IU/L	61.3 (76.8)	45.8 (34.7)	25.0 (8.9)	**0.0356**
C-peptide, ng/mL	7.07 (3.84)	6.36 (2.80)	4.47 (1.36)	**0.0150**
Glucose, mg/dL	152.4 (75.9)	123.9 (50.5)	106.7 (23.2)	**0.0216**
HbA1c (%)	6.65 (1.02)	6.43 (1.25)	6.12 (1.03)	0.2020
Adiponectin, µg/mL	3.894 (2.145)	3.982 (2.037)	5.221 (3.193)	0.1360
Lipid and lipoprotein metabolism				
Serum triglyceride, mg/dL	205.2 (123.6)	196.3 (124.8)	176.8 (75.8)	0.4936
HDL cholesterol, mg/dL	45.6 (10.6)	39.9 (9.8)	39.6 (11.4)	0.1096
Non-HDL-cholesterol, mg/dL	143.7 (35.2)	156.9 (58.2)	144.3 (50.7)	0.9552
ApoB, mg/dL	103.3 (22.5)	108.5 (34.5)	101.3 (29.2)	0.8650
ApoC-III, mg/dL	11.00 (5.54)	10.86 (7.09)	10.36 (4.26)	0.9514

*one-way ANOVA for trend, unadjusted p values

Carriers of PNPLA3 148M carriers presented with NASH of comparable severity to non-carriers at a younger age and with less baseline insulin resistance

Table 3. EFX improved NASH histology across PNPLA3 genotypes

BALANCED Study, F1-F3 NASH	# of PNPLA3 148M risk alleles		
	0 (I/I)	1 (I/M)	2 (M/M)
EFX-treated patients with end-of-study biopsy, N	**10**	**18**	**9**
NASH resolution, n (%)	4 (40%)	9 (50%)	5 (56%)
≥4-point improvement in NAS	5 (50%)	8 (44%)	5 (56%)
Ballooning resolution, n (%)	4 (40%)	9 (50%)	9 (100%)
≥1-stage improvement in fibrosis, n (%)	4 (40%)	10 (56%)	7 (78%)
2-stage improvement in fibrosis in patients with F2/F3 at baseline, n/N (%)	3/6 (50%)	4/10 (40%)	4/5 (80%)
≥1-stage improvement in fibrosis AND NASH resolution, n (%)	2 (20%)	5 (28%)	4 (44%)

Only two patients receiving placebo achieved ≥30% relative reduction in liver fat content and received an end-of-treatment biopsy, preventing meaningful comparison across genotypes in the placebo group

Figure 2. EFX improved markers of liver injury (A, ALT; B, AST; C, GGT) and fibrogenesis (D, Pro-C3) across PNPLA3 genotypes

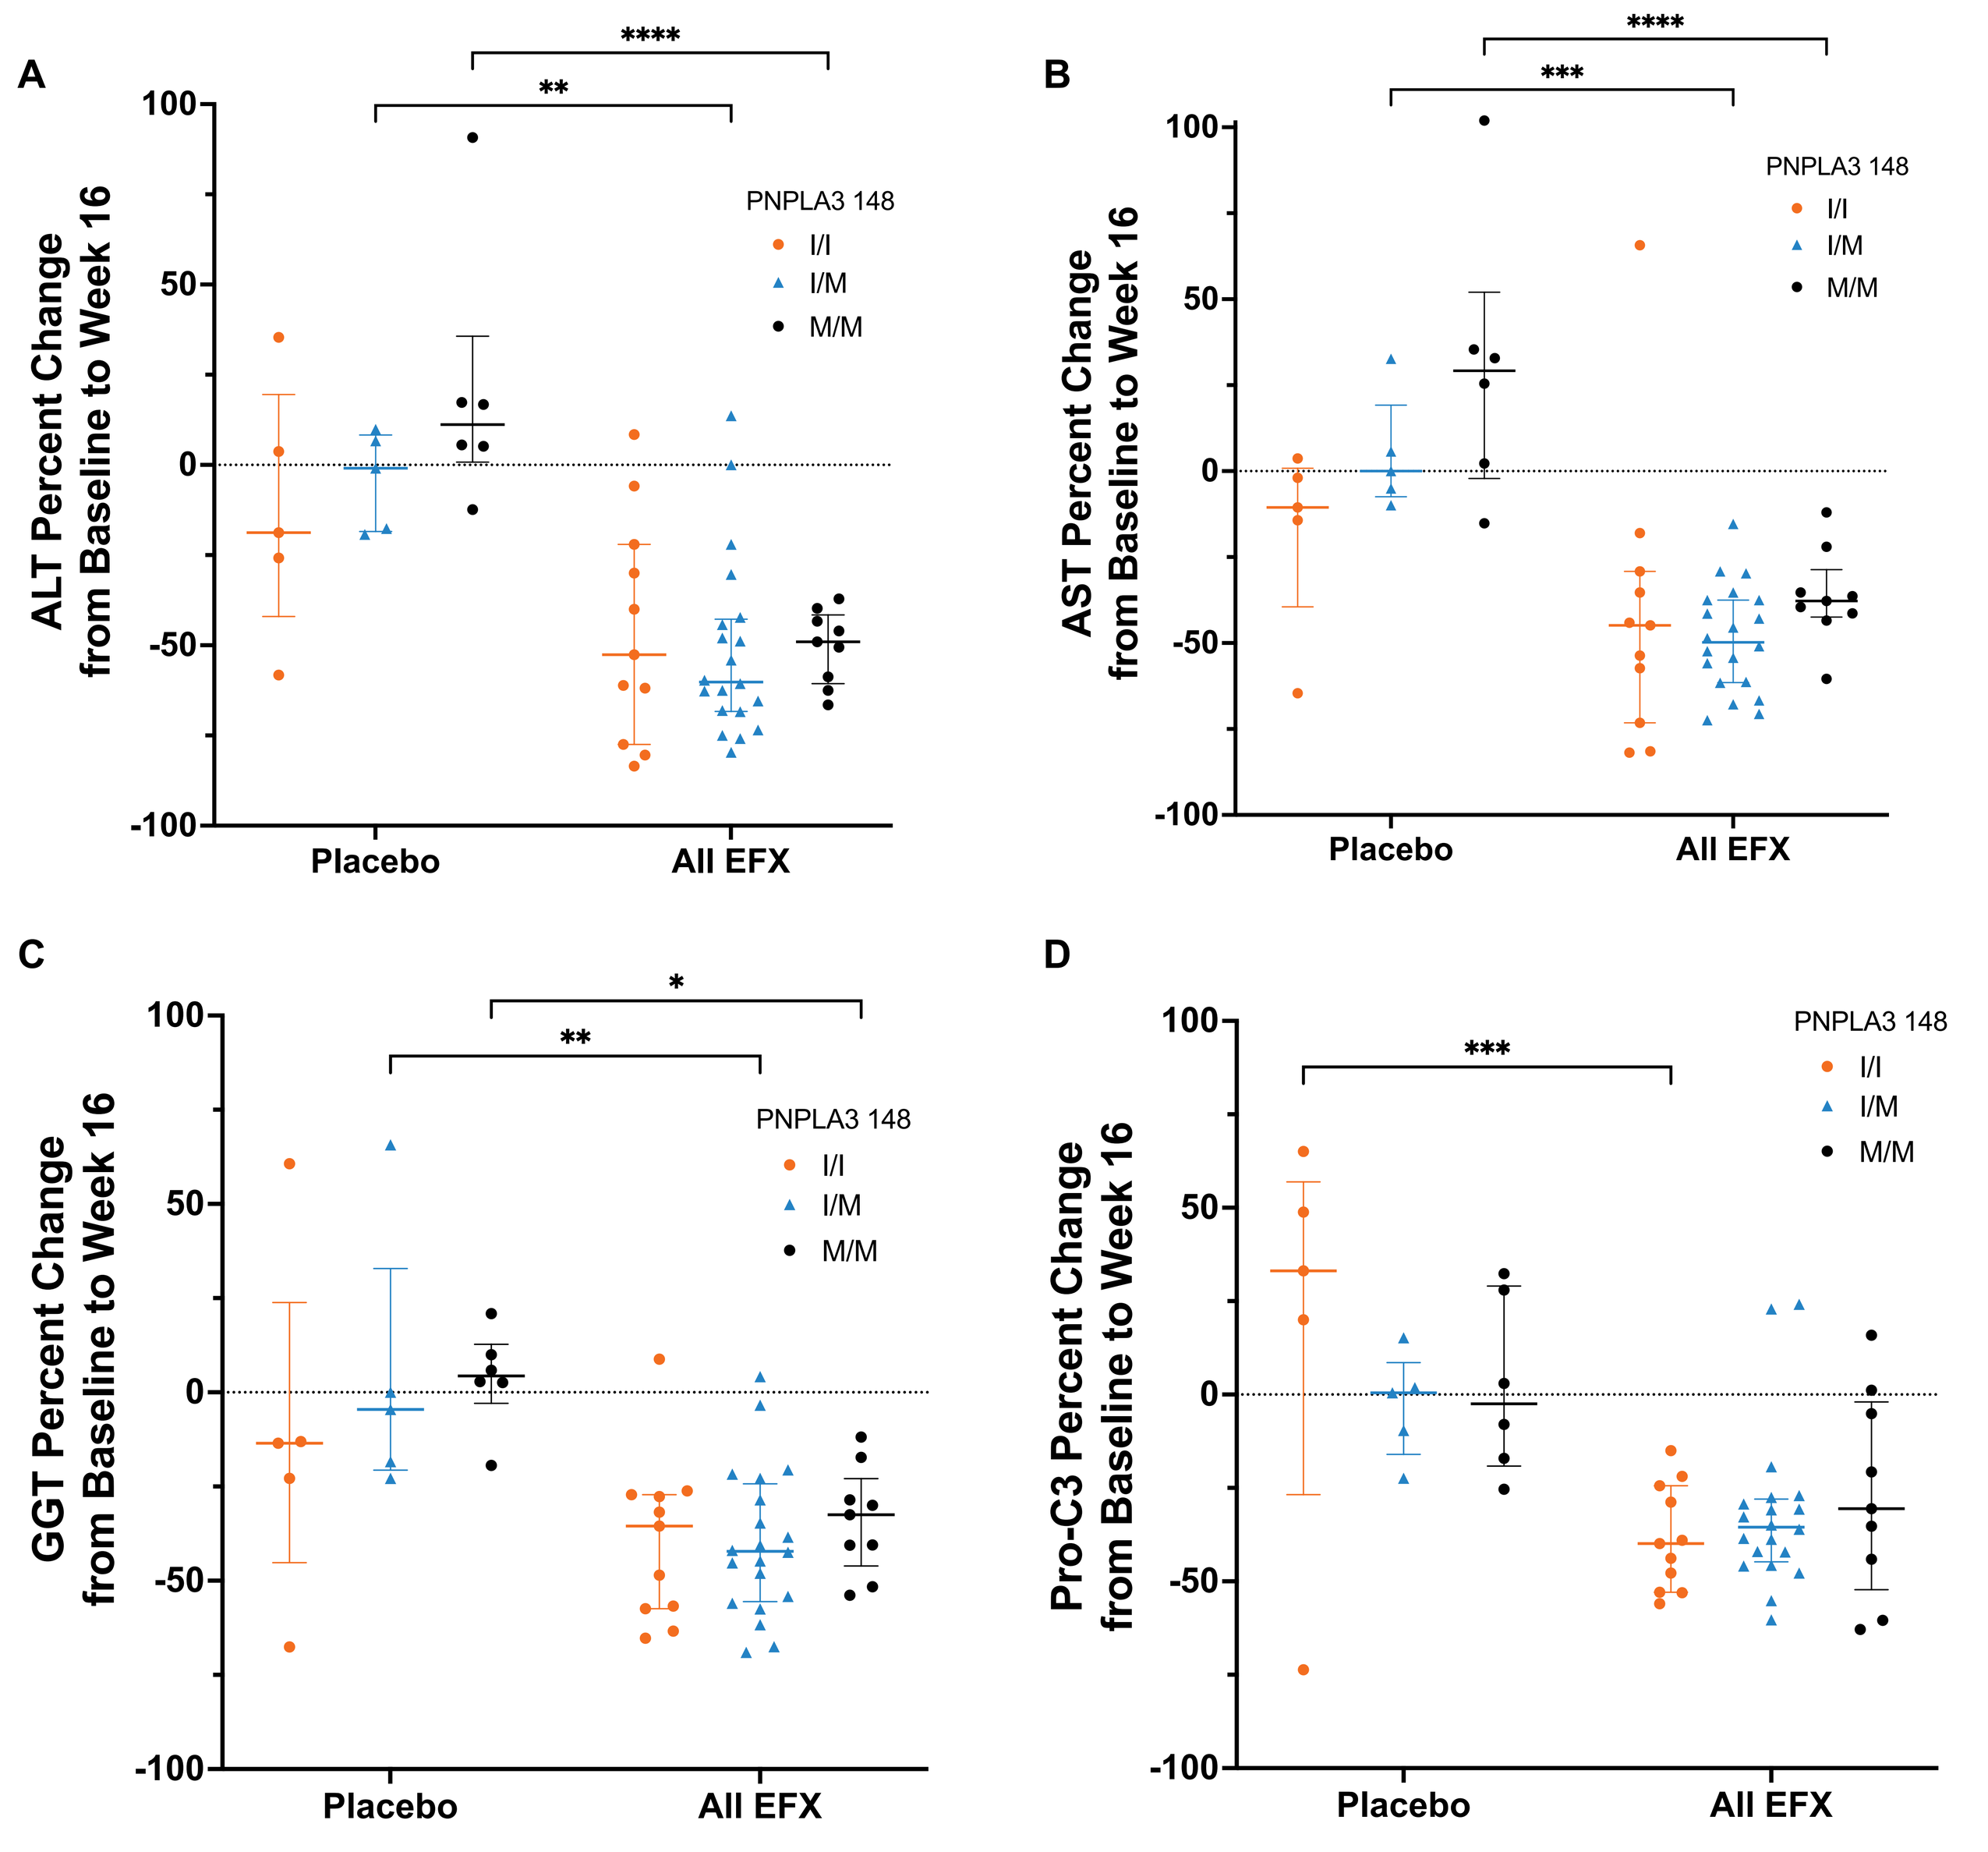

A
ALT Percent Change from Baseline to Week 16
B
AST Percent Change from Baseline to Week 16
C
GGT Percent Change from Baseline to Week 16
D
Pro-C3 Percent Change from Baseline to Week 16
PNPLA3 148
I/I
I/M
M/M
Placebo
All EFX

* p<0.05, ** p<0.01, *** p<0.001, **** p<0.0001 (two-way ANOVA, Sidak's post hoc test)

Figure 3. EFX improved liver fat content (A) and serum triglyceride (B) across PNPLA3 genotypes

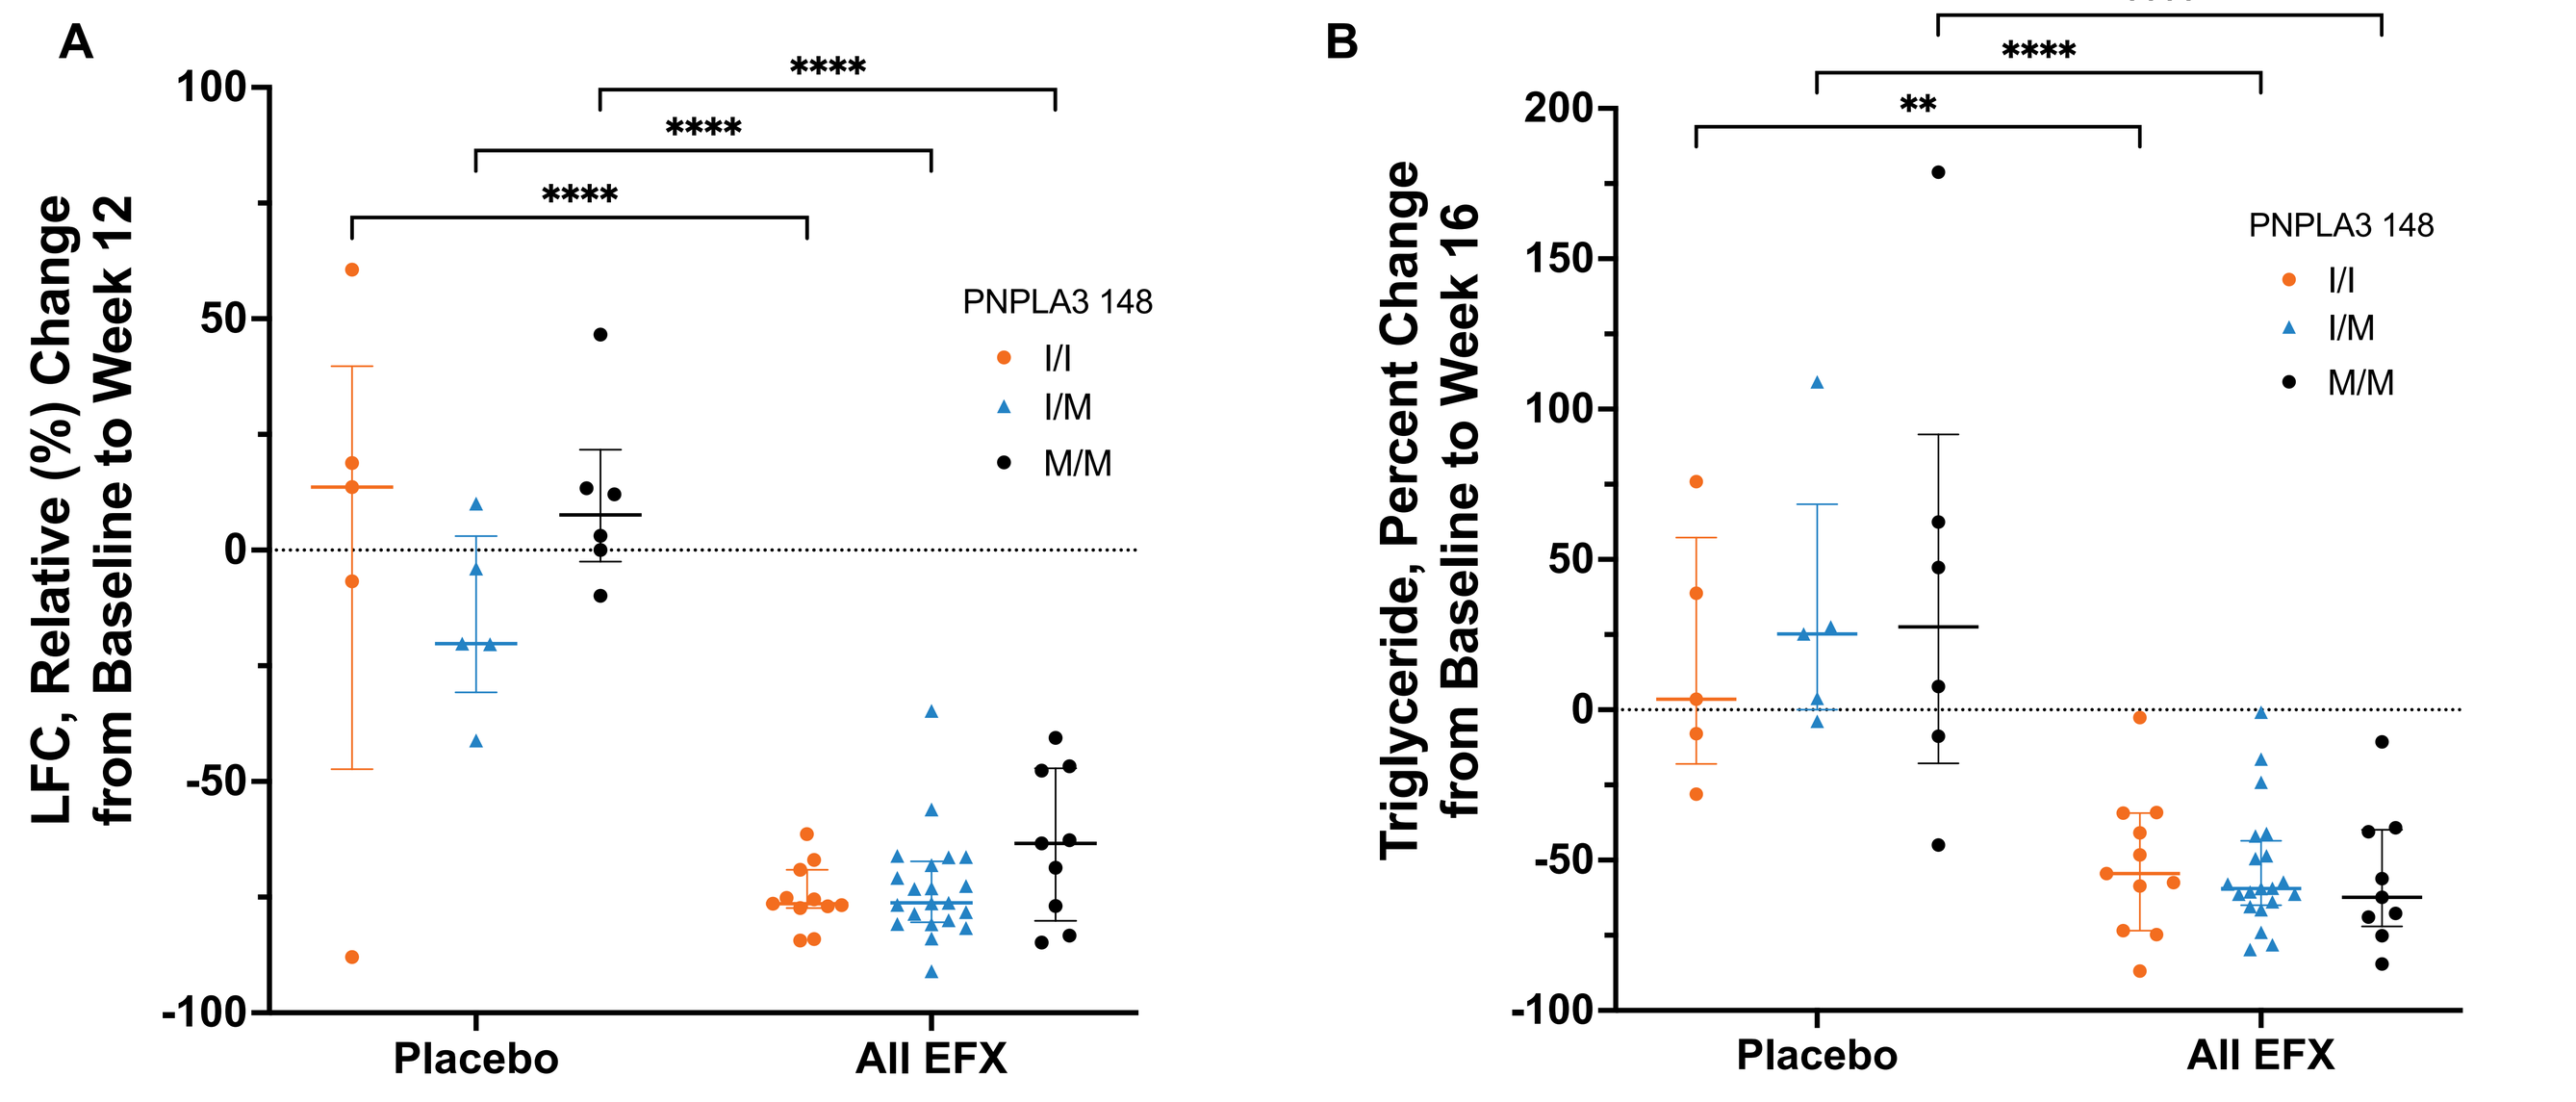

A
LFC, Relative (%) Change from Baseline to Week 12
B
Triglyceride, Percent Change from Baseline to Week 16
PNPLA3 148
I/I
I/M
M/M
Placebo
All EFX

** p<0.01, **** p<0.0001 (two-way ANOVA, Sidak's post hoc test)

Figure 4. EFX improved markers of glucose metabolism and insulin sensitivity across PNPLA3 genotypes (A, HbA1c; B, C-peptide; C, Adiponectin; D, HOMA-IR)

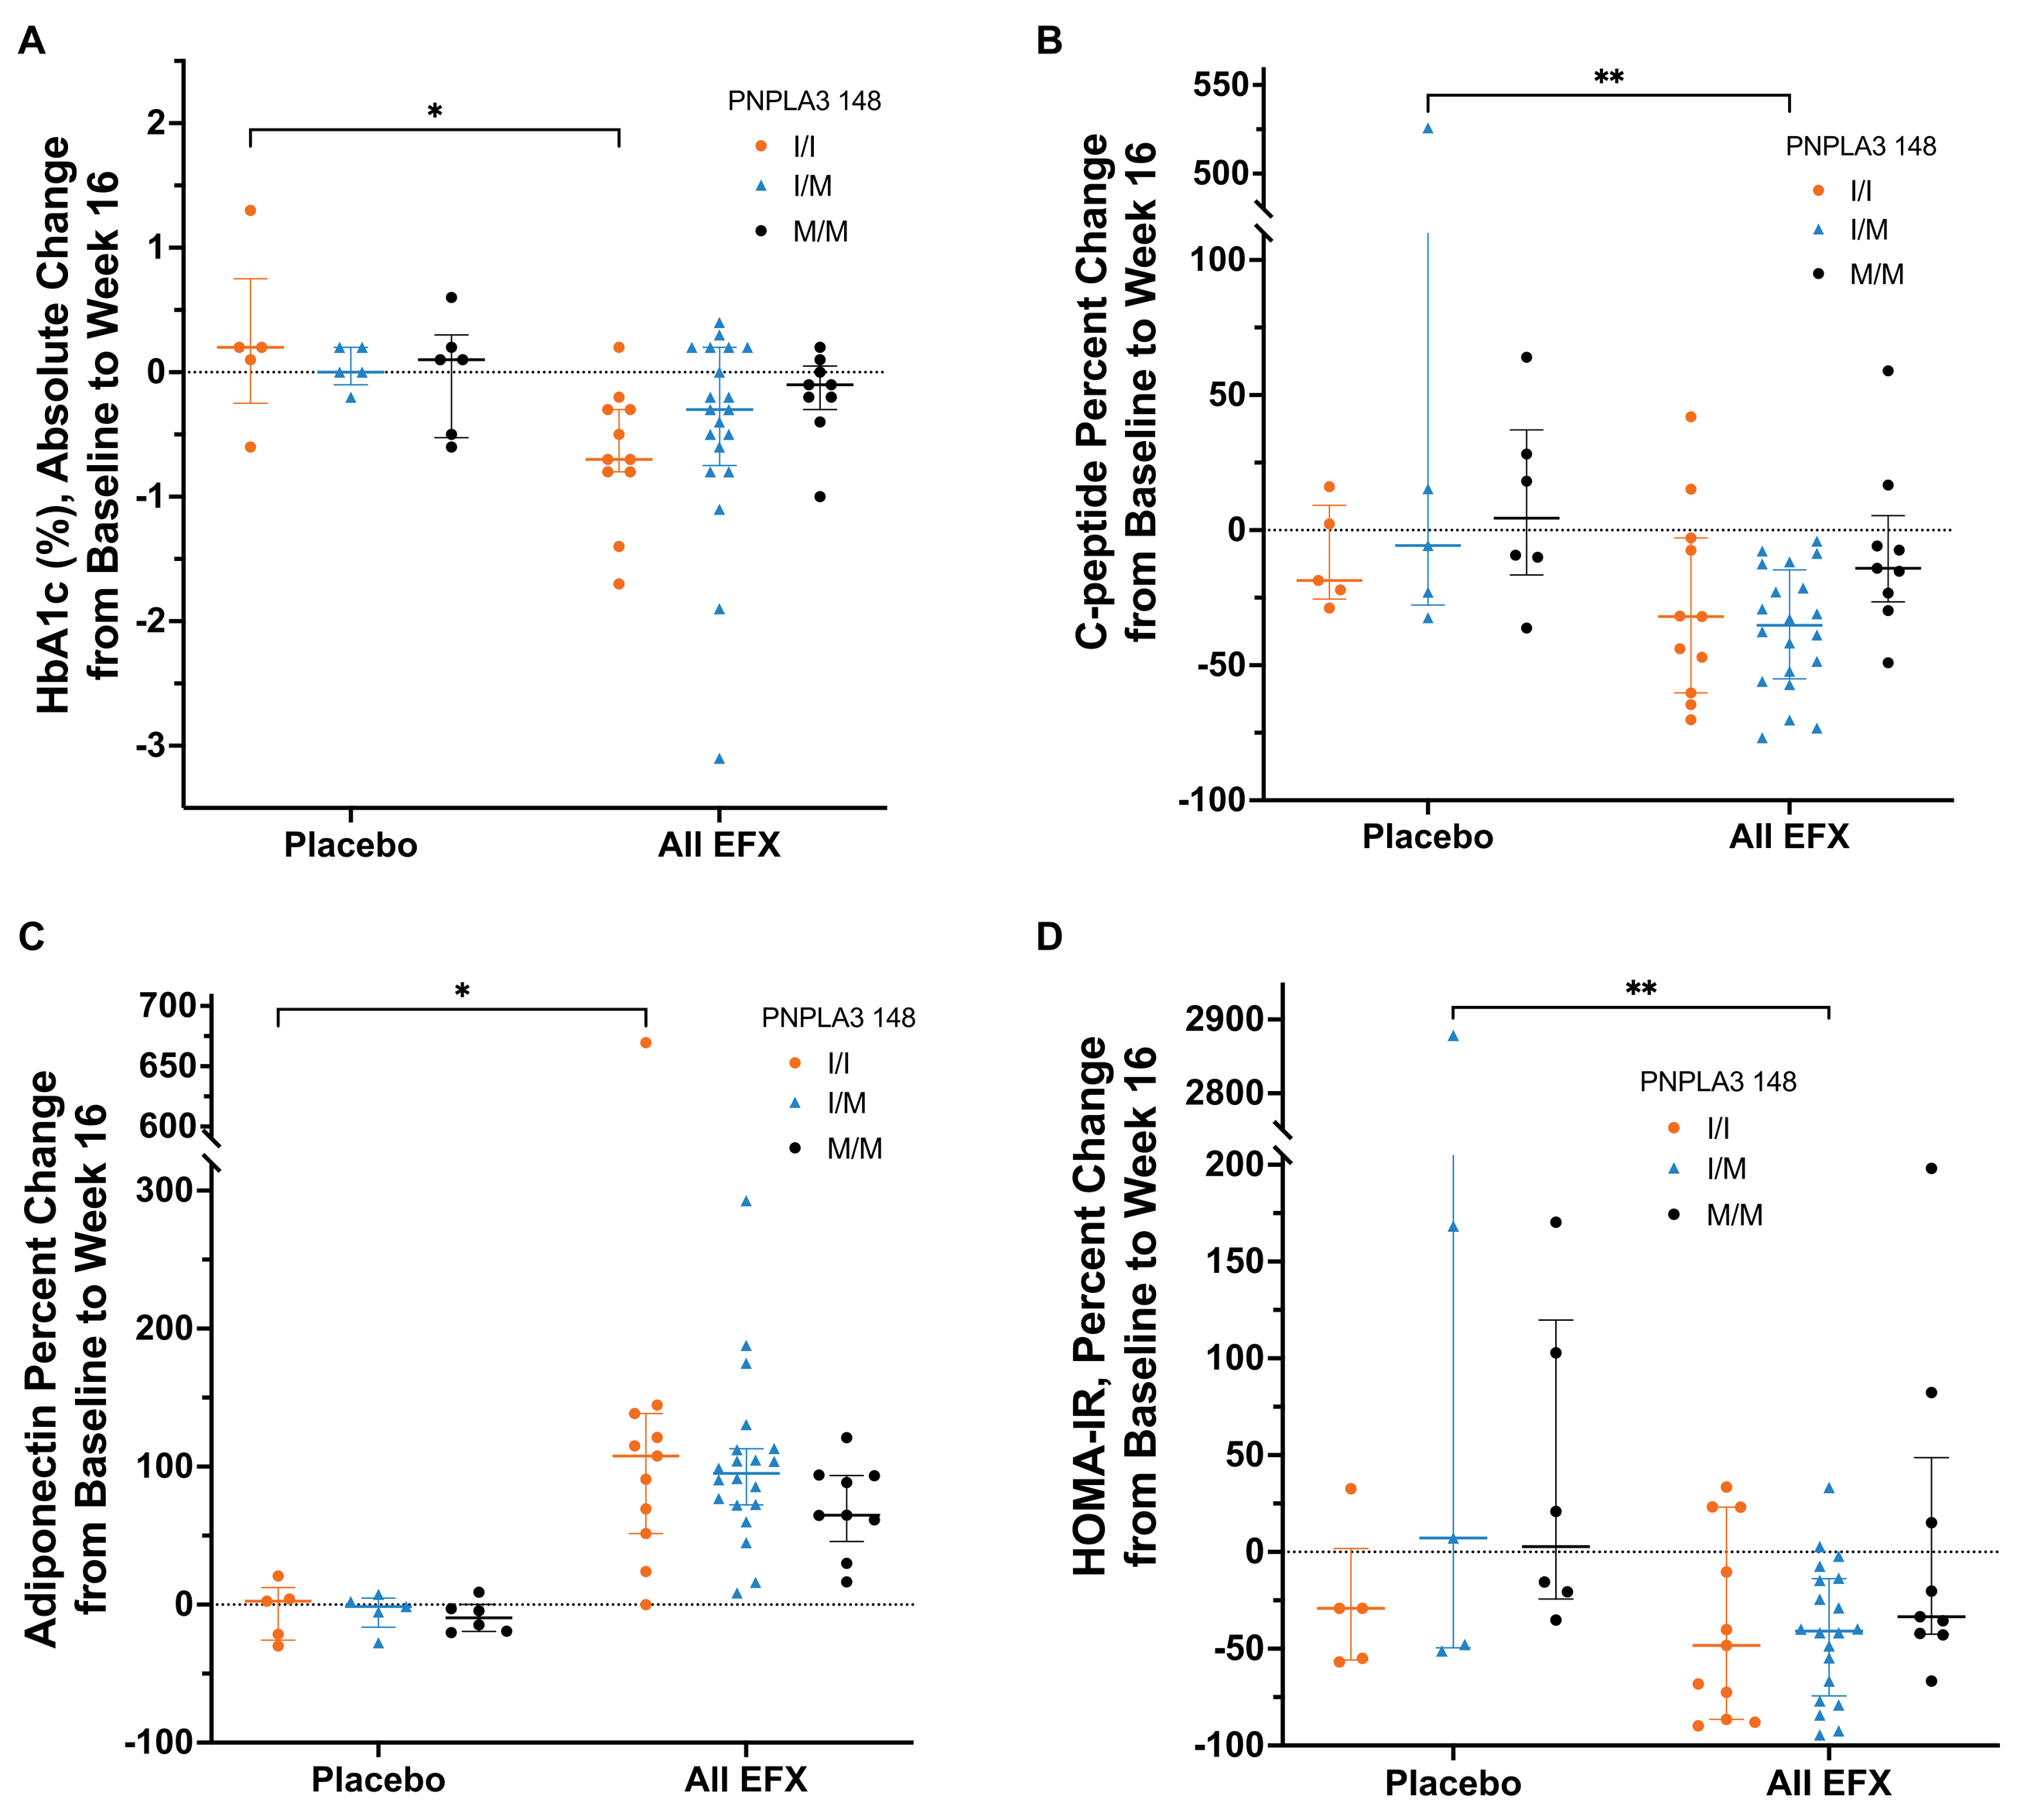

A
HbA1c (%), Absolute Change from Baseline to Week 16
B
C-peptide Percent Change from Baseline to Week 16
C
Adiponectin Percent Change from Baseline to Week 16
D
HOMA-IR, Percent Change from Baseline to Week 16
PNPLA3 148
I/I
I/M
M/M
Placebo
All EFX

* p<0.05, ** p<0.01 (two-way ANOVA, Sidak's post hoc test)

All graphs show median ± IQR

CONCLUSIONS

- In BALANCED, patients genetically predisposed to progressive disease, *i.e.*, PNPLA3 I148M carriers, presented with NASH of comparable severity, but were younger and less insulin resistant than non-carriers
- EFX improved components of NASH histology and fibrosis across *PNPLA3* genotypes, notably at least maintaining effects in those carrying I148M, including 100% of M/M patients achieving ballooning resolution and 80% of F2/F3 patients achieving 2-stage fibrosis improvement
- EFX reduced markers of liver injury across PNPLA3 genotypes, notably among M/M homozygotes, for whom these markers tended to worsen when untreated over 16 weeks
- EFX tended to reduce Pro-C3 across PNPLA3 genotypes, which was more variable in its response
- EFX consistently and robustly reduced liver fat and serum triglyceride across PNPLA3 genotypes
- **EFX improved metabolic health of liver both in patients whose NASH carries a significant genetic component, but also in patients whose NASH appears to be driven primarily by metabolic dysfunction, *e.g.* T2D**
- Consistent with their more severe metabolic dysfunction and insulin resistance at baseline, I/I and I/M patients achieved greatest improvements in insulin sensitivity and glycaemic control, though metabolic markers tended to improve even among the M/M patients with less metabolic dysfunction

Histological improvements were maintained in patients at highest risk of progression to ESLD.

EFX significantly improved liver manifestations of metabolic disease across PNPLA3 genotypes.

REFERENCES

1. Harrison *et al.* (2021) *Nat Med* 27:1262-71
2. Romeo *et al.* (2008) *Nat Genet* 40(12):1461-5
3. Barrata *et al.* (2019) *Heptaol Commun* 3:894-907

ACKNOWLEDGMENTS

The authors wish to thank the patients, their families, and study investigators. This study and all analyses were funded by Akero Therapeutics.